Targeted Delivery of Neoantigens to Antigen Presenting Cells using Vaccibodies Encoded in mRNA Results in Stronger and Broader T Cell Responses, and Improved Tumor Control

Brian T. Weinert[1], Lise Skullerud[1], Eirik Solbakken[1], Ana Textor[1], Mirjam Dürkoop[1], Inga Patrycja Winge[1], Gunnstein Norheim[1], Elisabeth Stubsrud[1], Stine Granum[1], Mikkel W. Pedersen[1], Håkan Norell[1]

[1]Nykode Therapeutics ASA, Oslo, Norway

nykode therapeutics

Background

Individualized cancer vaccines based on tumor-specific neoantigens is a groundbreaking approach in oncology that has shown promising efficacy in recent clinical trials. Nykode Therapeutics developed a vaccine platform (VaccibodyTM) that targets antigens directly to antigen presenting cells (APCs). Here we used a model Vaccibody modular fusion protein containing Chemokine ligand 3-like 1 (CCL3L1) to bind and attract APCs, a dimerization domain to facilitate strong bivalent binding, and an antigen unit encoding 20 neoantigens. Individualized neoantigen vaccines delivered in a DNA-encoded Vaccibody induce strong and broad T cell responses in pre-clinical and clinical settings.

In the study presented here, we compared the immunogenicity and tumor control of a mRNA-encoded Vaccibody vaccine to an untargeted antigen vaccine. Both vaccines encoded the same 20 neoantigens derived from the murine MC38 tumor model and were administered in a lipid nanoparticle (LNP) format.

Find out more about our neoantigen vaccines at our website:
https://nykode.com/research-and-development/scientific-papers-and-presentations/

The Vaccibody platform

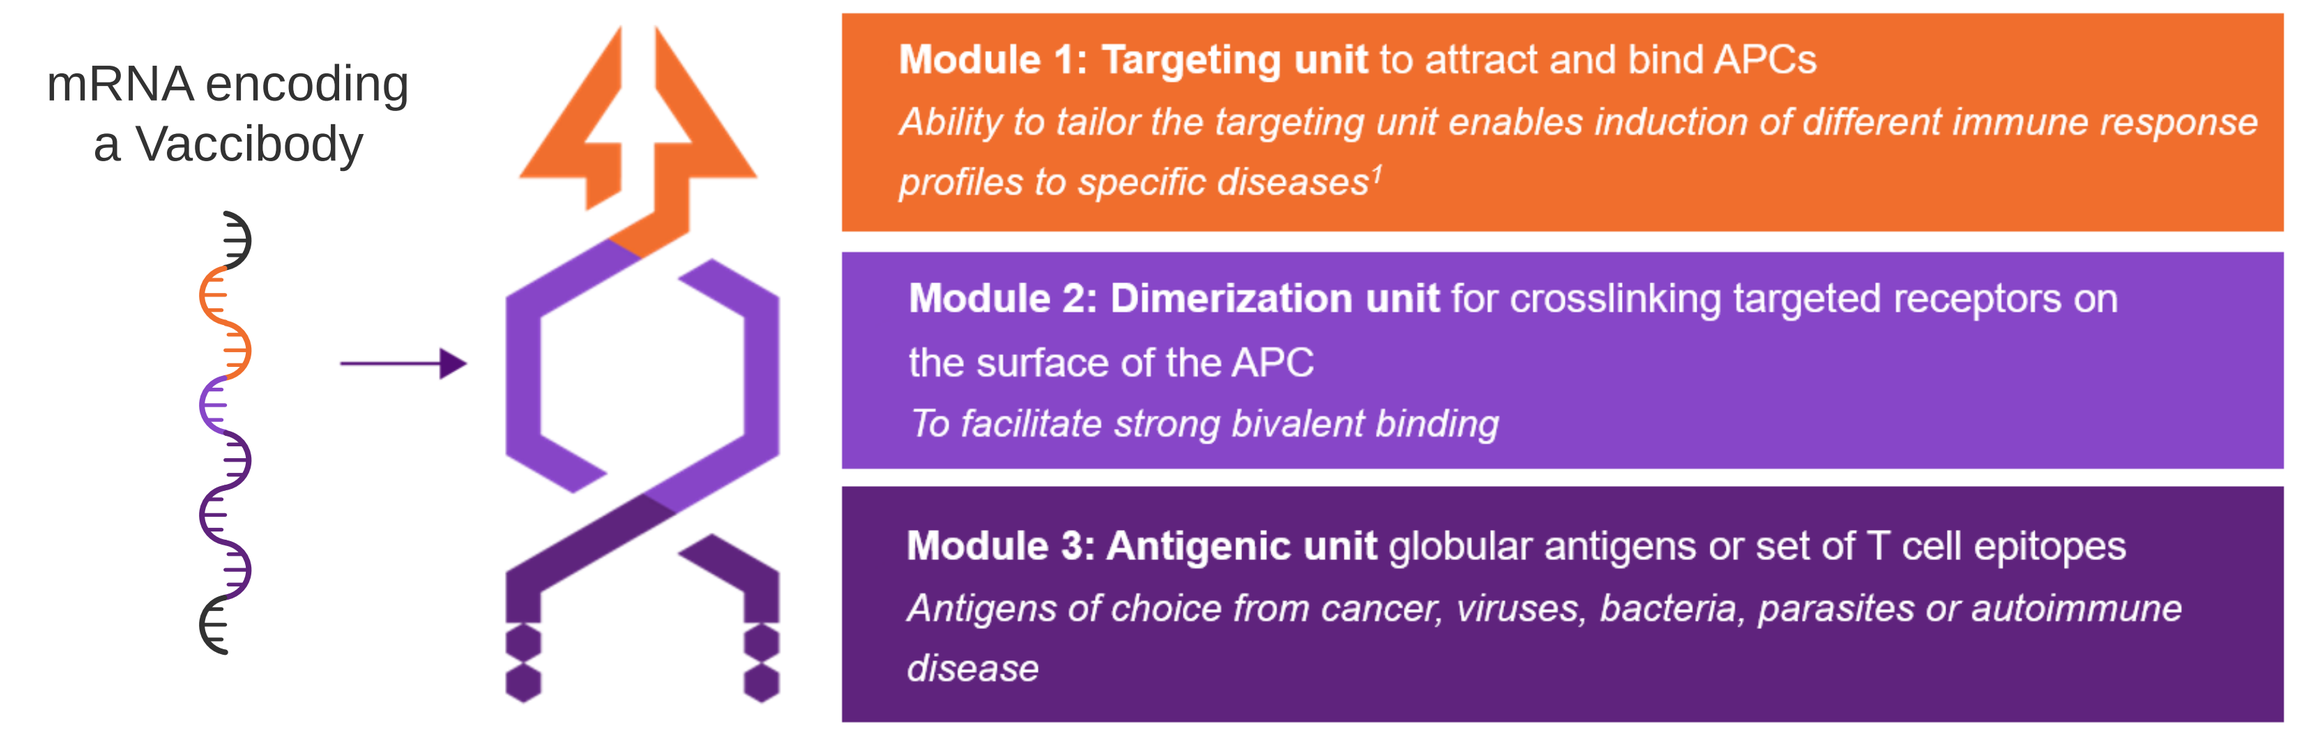
mRNA encoding a Vaccibody
Module 1: Targeting unit to attract and bind APCs
Ability to tailor the targeting unit enables induction of different immune response profiles to specific diseases[1]
Module 2: Dimerization unit for crosslinking targeted receptors on the surface of the APC
To facilitate strong bivalent binding
Module 3: Antigenic unit globular antigens or set of T cell epitopes
Antigens of choice from cancer, viruses, bacteria, parasites or autoimmune disease

Vaccibody-based immunotherapies may be delivered via DNA, mRNA, viral vectors or as recombinant proteins

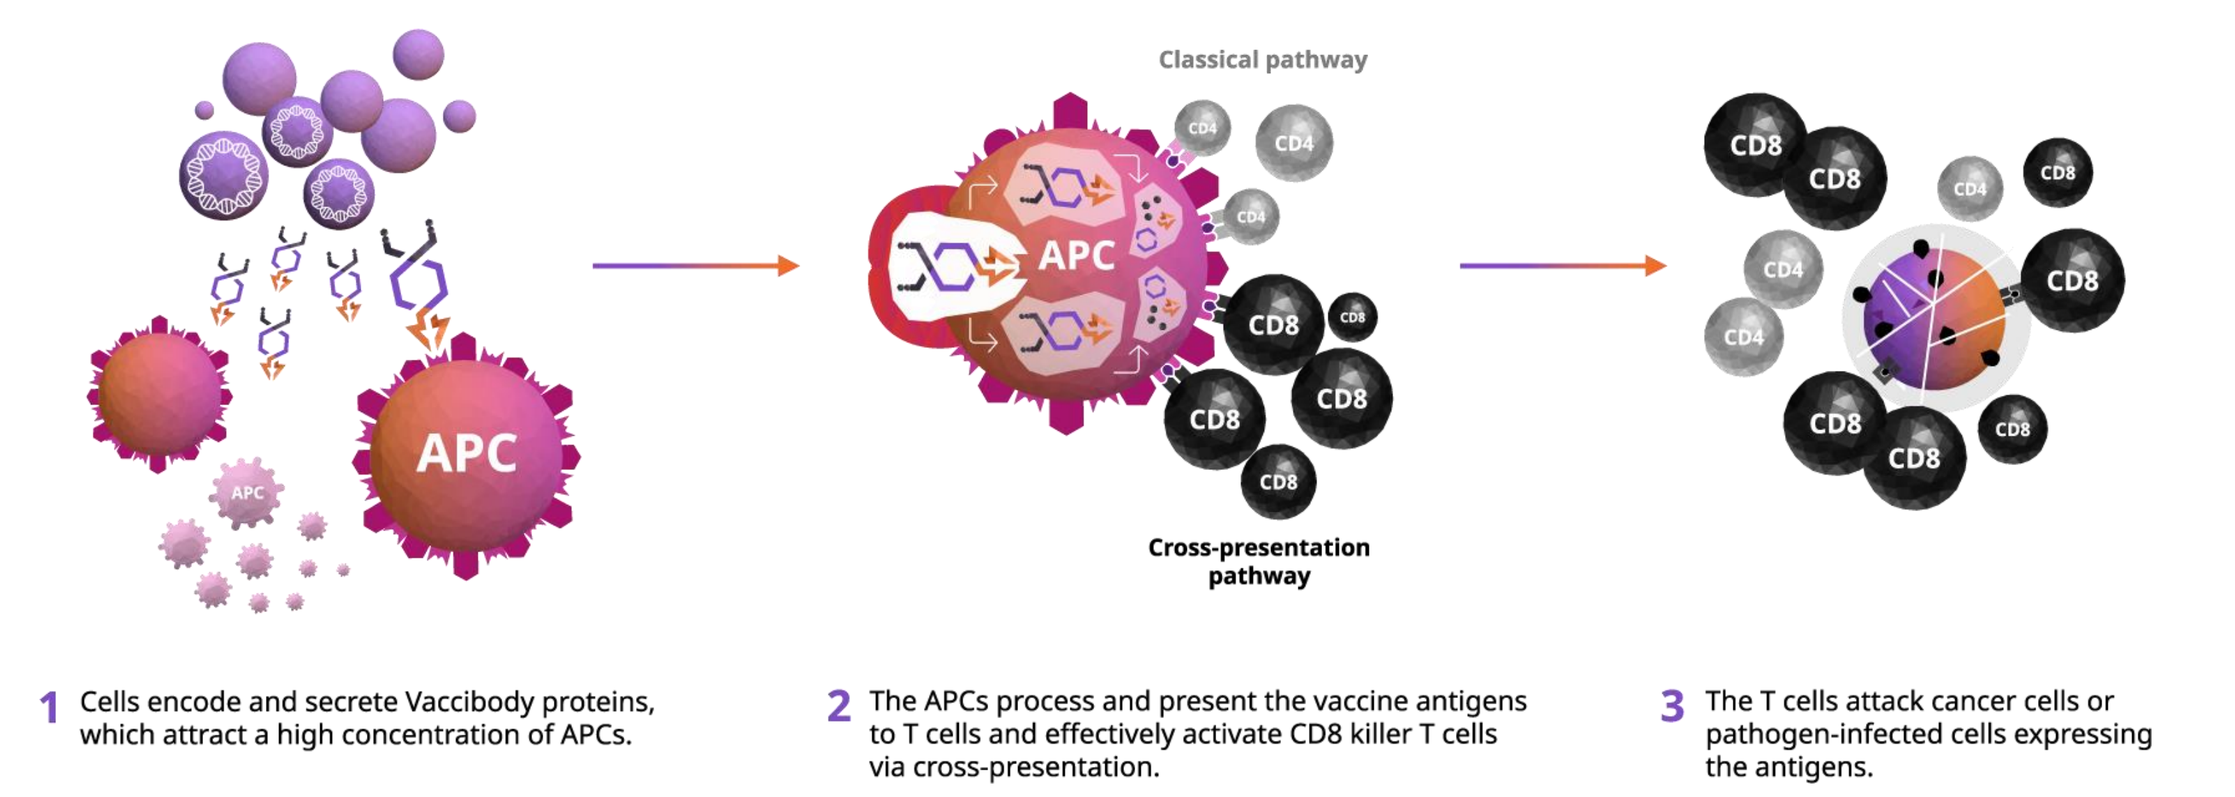
APC
Classical pathway
Cross-presentation pathway
CD8
CD4
1 Cells encode and secrete Vaccibody proteins, which attract a high concentration of APCs.
2 The APCs process and present the vaccine antigens to T cells and effectively activate CD8 killer T cells via cross-presentation.
3 The T cells attack cancer cells or pathogen-infected cells expressing the antigens.

Vaccibody neoantigen vaccines induce stronger and broader T cell responses

Vaccine constructs & delivery vectors used in these studies

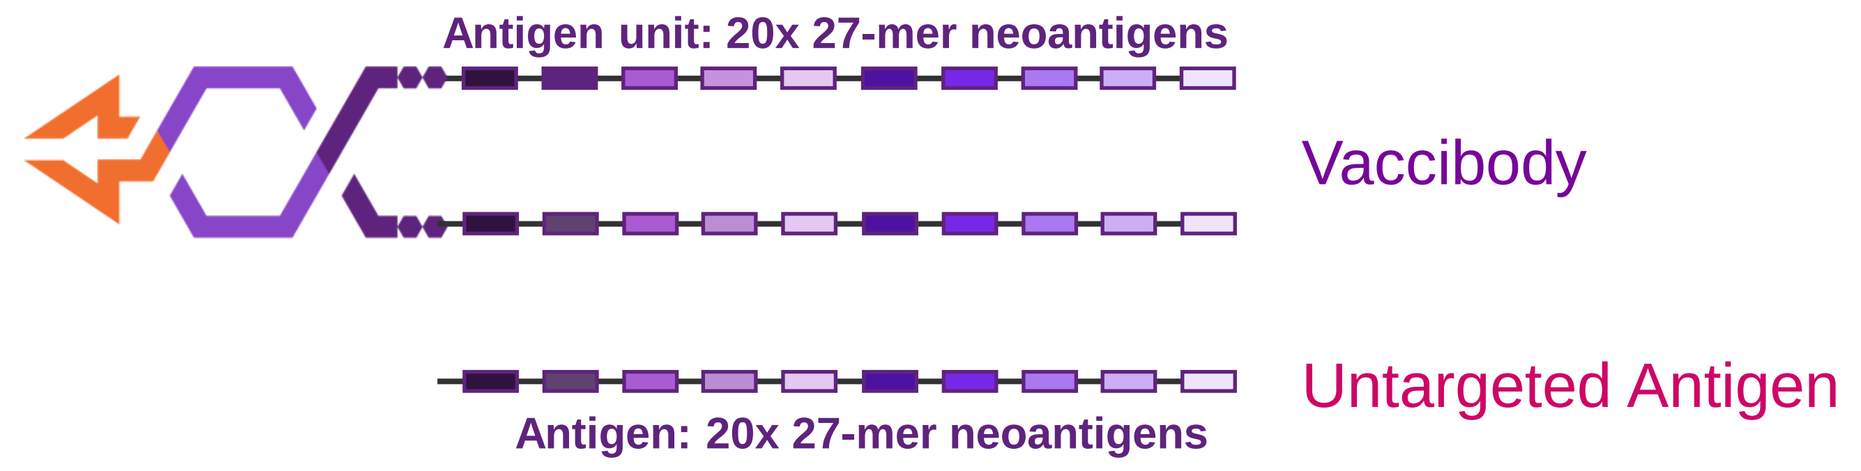
Antigen unit: 20x 27-mer neoantigens
Vaccibody
Untargeted Antigen
Antigen: 20x 27-mer neoantigens

Vaccines were administered as mRNA formulated in lipid nanoparticles (LNPs) and injected intramuscularly.

The Vaccibody neoantigen vaccine elicited stronger IFNγ T cell responses than the untargeted neoantigen vaccine

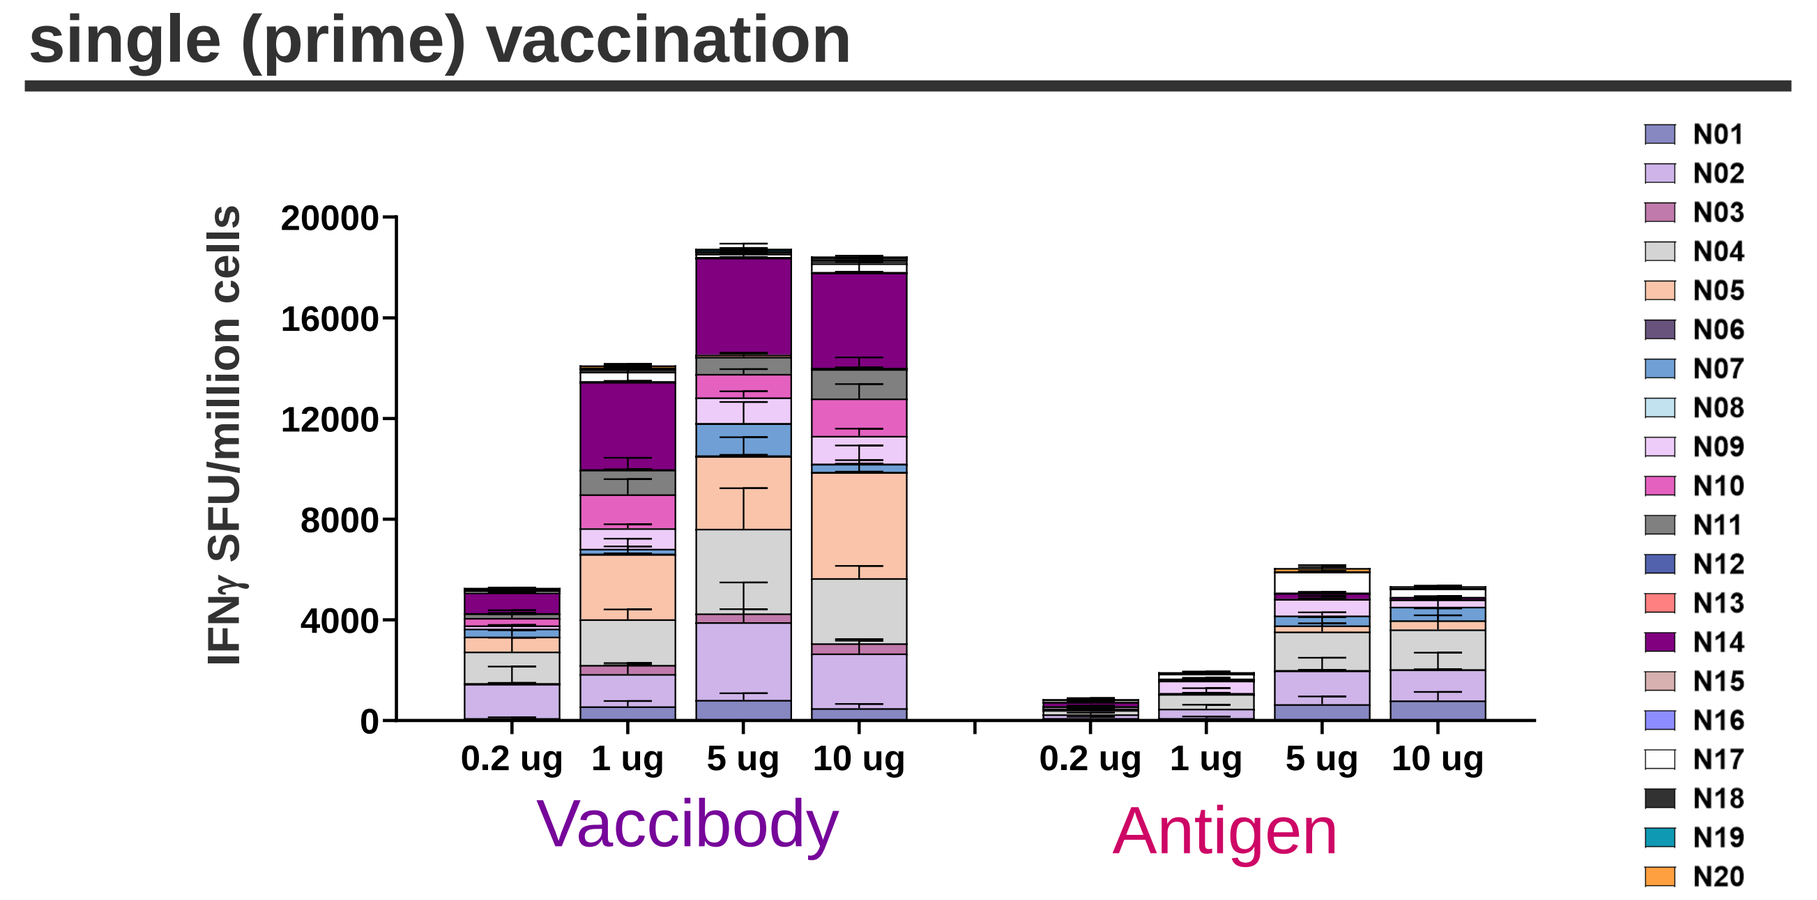
single (prime) vaccination
IFNγ SFU/million cells
0.2 ug
1 ug
5 ug
10 ug
Vaccibody
Antigen
N01 N02 N03 N04 N05 N06 N07 N08 N09 N10 N11 N12 N13 N14 N15 N16 N17 N18 N19 N20

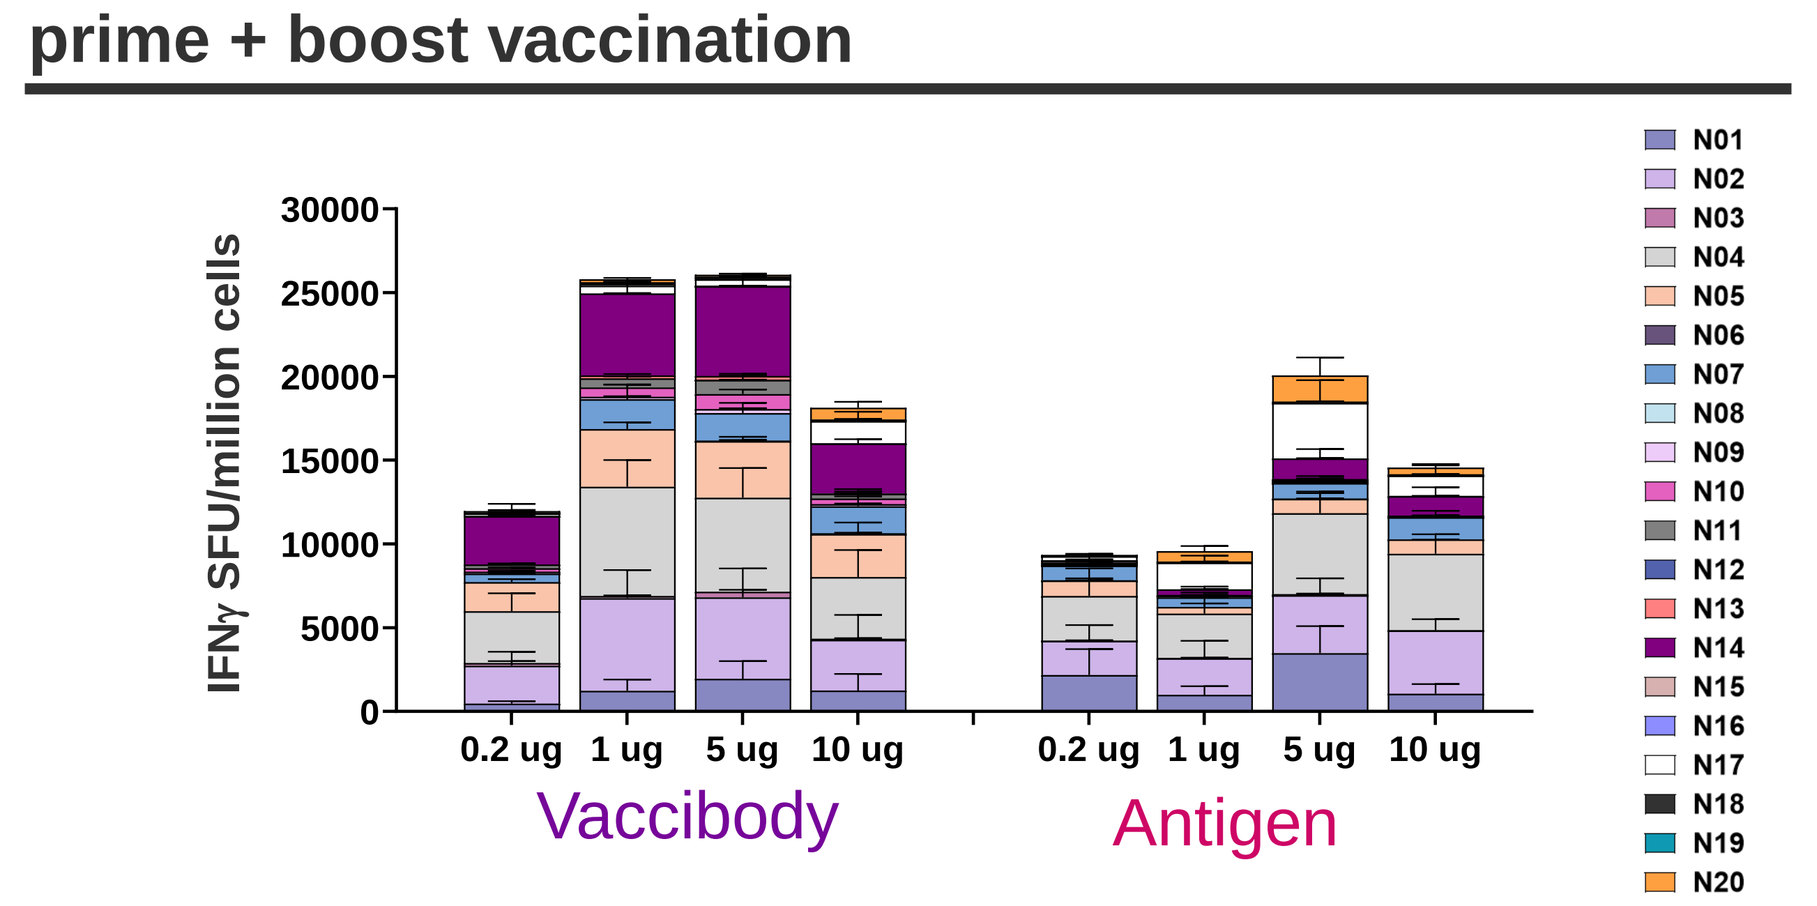
prime + boost vaccination
IFNγ SFU/million cells
0.2 ug
1 ug
5 ug
10 ug
Vaccibody
Antigen
N01 N02 N03 N04 N05 N06 N07 N08 N09 N10 N11 N12 N13 N14 N15 N16 N17 N18 N19 N20

The Vaccibody vaccine elicited a much stronger response (up to 7.5x stronger) than the antigen vaccine on day 7 following a single (prime) vaccination. After a second (boost) vaccination the Vaccibody vaccine still elicited a stronger response, although the magnitude of this difference was reduced. Note that the Vaccibody vaccine reached a peak response at a lower dose (1ug) than the antigen vaccine, highlighting the increased potency.

C57/BL6, n = 5 per group, IFNγ fluorospot assay of splenocytes using peptide pools covering each of the 20 neoantigens individually.

Vaccibody vaccines elicited responses to a greater number of neoantigens compared to the untargeted antigen vaccine

Average IFNγ SFU/million cells (prime + boost)

Neoantigen ID	Vaccibody 0.2 ug	Vaccibody 1 ug	Vaccibody 5 ug	Vaccibody 10 ug	Antigen 0.2 ug	Antigen 1 ug	Antigen 5 ug	Antigen 10 ug	
N01	465	1229	1960	1249	2178	990	3482	1055	
N02	2261	5538	4851	3026	2030	2193	3458	3784	
N03	174	121	340	-	-	-	-	-	*
N04	3078	6533	5610	3680	2662	2643	4823	4562	
N05	1739	3428	3373	2565	933	395	868	862	
N06	-	-	-	-	-	-	-	-	
N07	510	1793	1633	1617	886	568	934	1317	
N08	-	-	-	-	-	-	-	-	
N09	107	136	238	125	-	-	-	-	*
N10	207	566	902	335	-	-	-	-	*
N11	215	532	846	291	103	-	-	-	*
N12	-	-	-	-	-	-	-	-	
N13	-	176	228	-	-	-	-	-	*
N14	2871	4896	5354	2978	-	336	1214	1166	
N15	-	-	-	-	-	-	-	-	
N16	-	-	-	-	-	-	-	-	
N17	140	455	379	1319	241	1592	3301	1218	
N18	-	132	118	-	-	-	-	-	*
N19	-	-	-	-	-	-	-	-	
N20	-	182	104	699	-	629	1571	406	

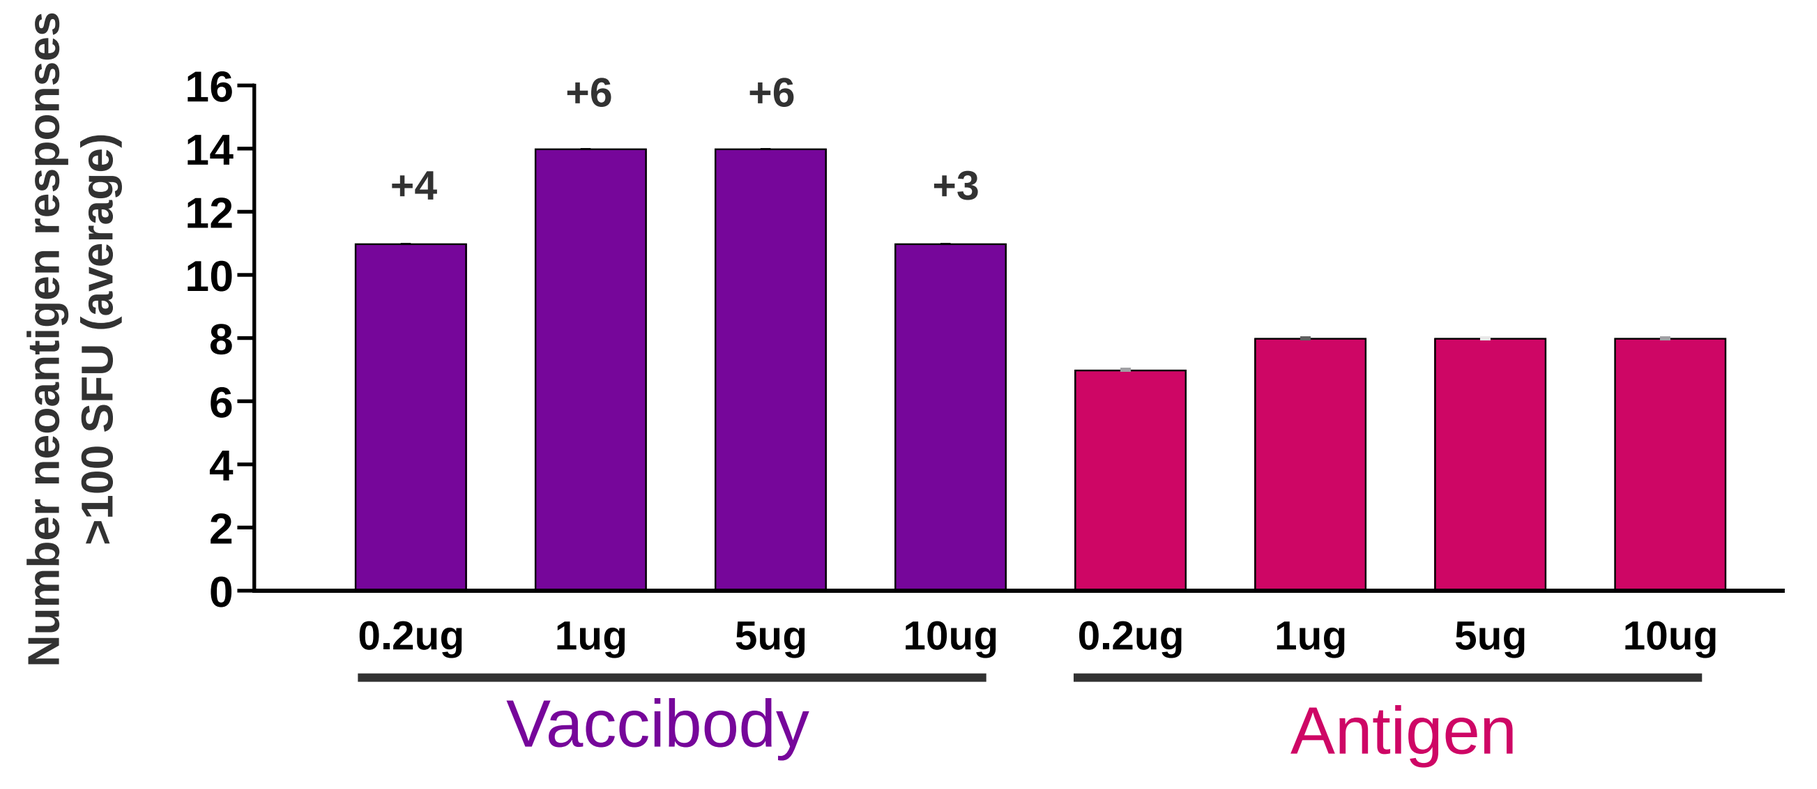
Number neoantigen responses >100 SFU (average)
+4
+6
+6
+3
0.2ug
1ug
5ug
10ug
Vaccibody
Antigen

The Vaccibody vaccine elicited responses to substantially more neoantigens than the untargeted neoantigen vaccine at the dose (5µg) that led to the strongest response in both vaccines after prime + boost vaccination.

The average interferon gamma (IFNγ) fluorospot signal is shown. Samples with less than 100 SFU are considered negative and are not shown.
() indicates neoantigens that elicited responses in the Vaccibody (VB) vaccine, but not in the unmodified antigen (AG) vaccine.*

Vaccibody neoantigen vaccines mediate improved tumor control

The Vaccibody neoantigen vaccine elicited far superior tumor control compared to the untargeted neoantigen vaccine

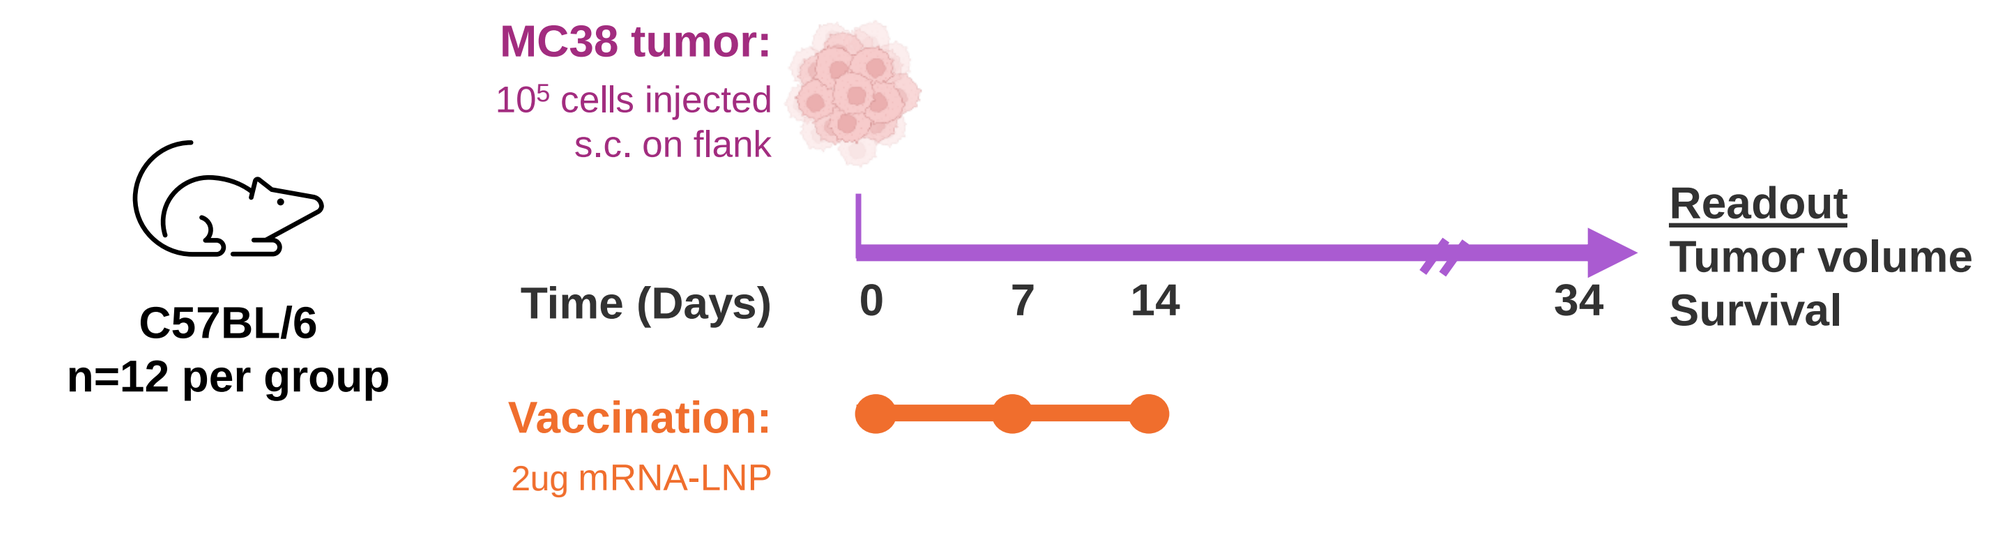
C57BL/6
n=12 per group
MC38 tumor:
10^5 cells injected s.c. on flank
Time (Days)
0
7
14
34
Readout
Tumor volume
Survival
Vaccination:
2ug mRNA-LNP

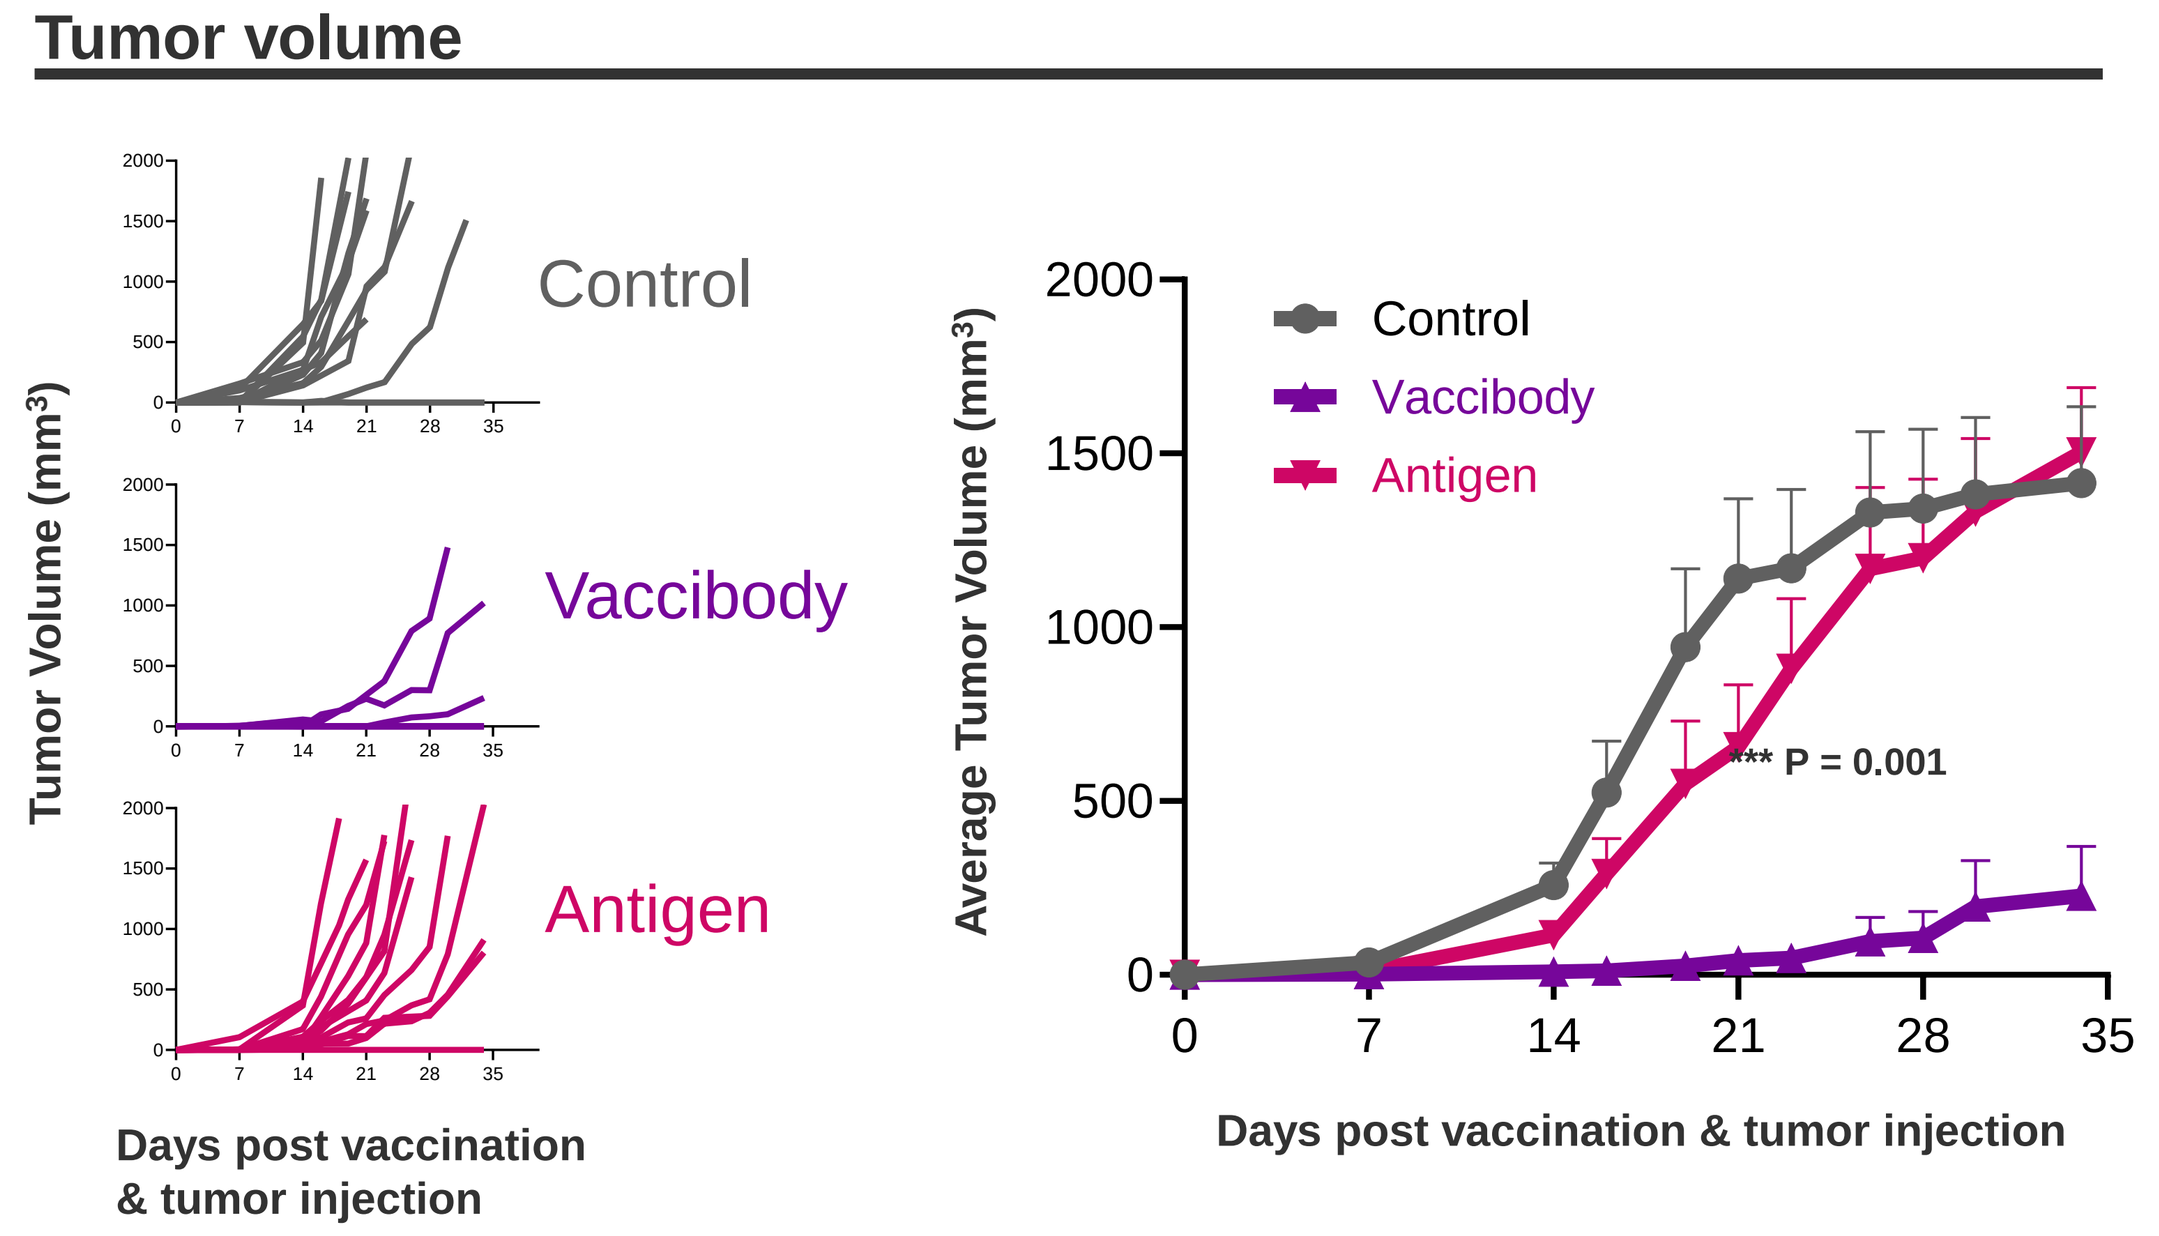
Tumor volume
Tumor Volume (mm³)
Control
Vaccibody
Antigen
Days post vaccination & tumor injection
Average Tumor Volume (mm³)
*** P = 0.001
Days post vaccination & tumor injection

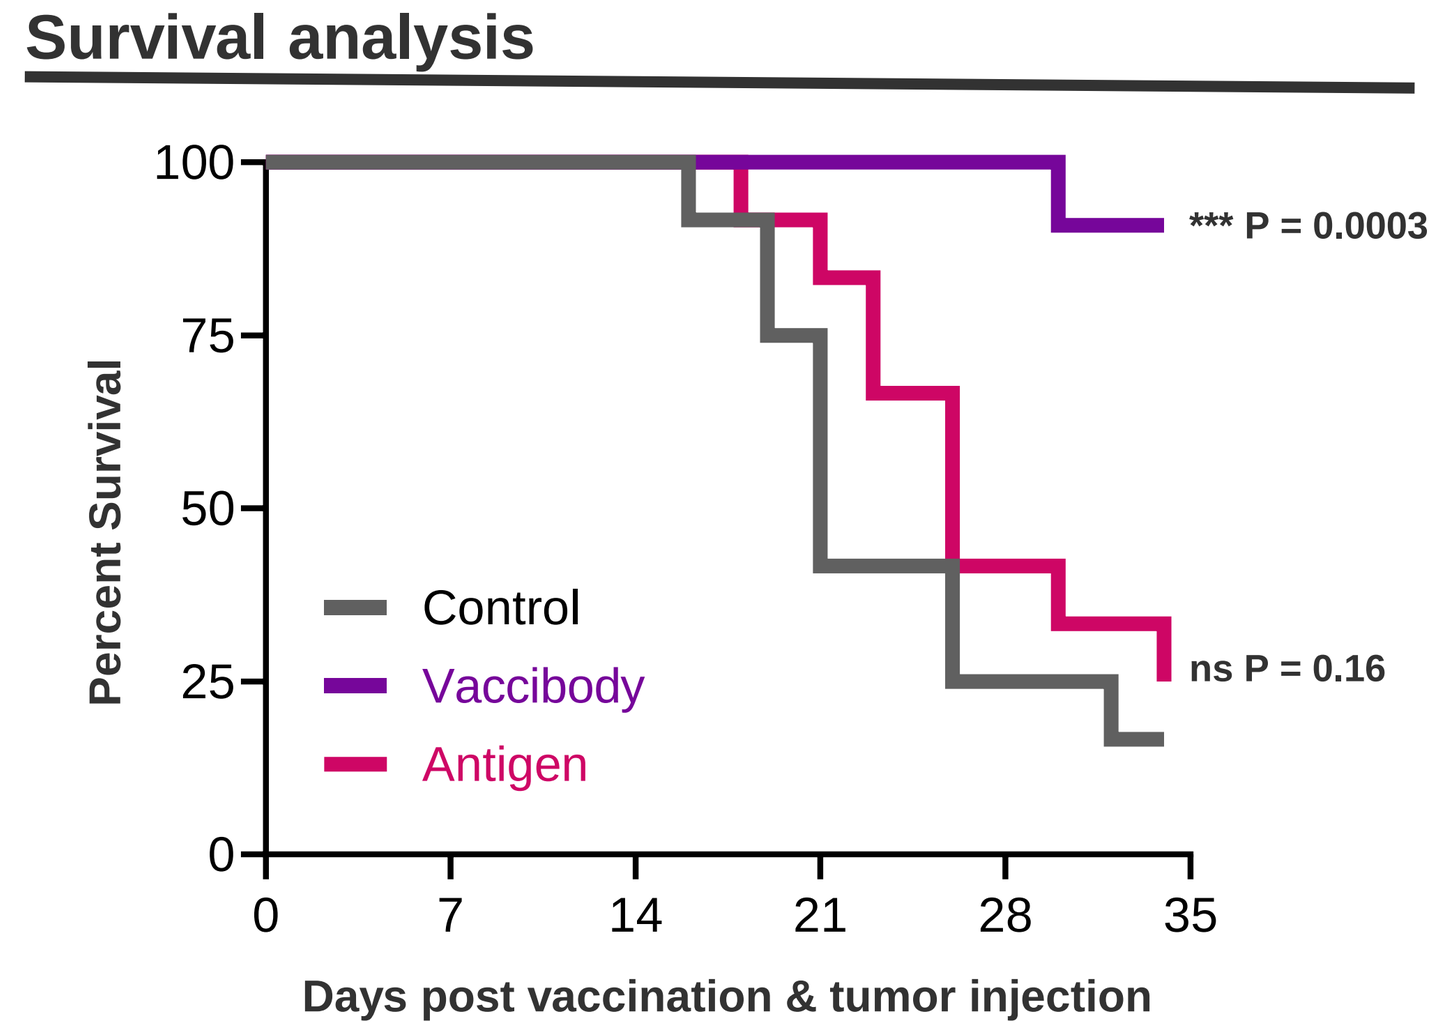
Survival analysis
Percent Survival
*** P = 0.0003
ns P = 0.16
Control
Vaccibody
Antigen
Days post vaccination & tumor injection

Tumor rejection

Group	Tumors Rejected	P-value (Fisher test)
Control	2/12	NA
Vaccibody	8/11	0.01
Antigen	1/12	1.00

The Vaccibody vaccine led to significantly enhanced tumor rejection (Fisher), significantly reduced tumor growth (P=0.001, Tukey), significantly increased survival (Gehan-Breslow-Wilcoxon test), while the untargeted Antigen vaccine displayed no significant differences compared to control (LNP buffer) vaccine.

Conclusions

► **The Nykode Vaccibody platform, when delivered in a mRNA-LNP format, elicits stronger and broader T cell responses compared to an untargeted neoantigen vaccine.**

► **The Vaccibody neoantigen vaccine elicited T cell responses to substantially more neoantigens than the untargeted neoantigen vaccine (14 vs 8 at peak dose).**

► **The Vaccibody neoantigen vaccine provided far better tumor control than the untargeted neoantigen vaccine, with tumor rejection in 72% of animals compared to just 8% of animals.**

► **Vaccibody neoantigen vaccination resulted in a substantially higher survival rate at day 34 (91% for Vaccibody vs 25% for antigen).**

Presented at Nature Conferences-Cancer Immunotherapy: From bench to bedside and back, Boston, June 10, 2024. | Nykode Therapeutics ASA